RYZ101 (^{225}Ac-DOTATATE) + carboplatin + etoposide + atezolizumab in somatostatin receptor expressing extensive-stage small-cell lung cancer

Sonam Puri[1], Wallace Akerley[1], Robert Langdon[2], Samuel Mehr[2], Noridza Rivera-Rodriguez[3], Jason Berilgen[4], Justin M. Rineer[5], Claire Mulvey[6], Krisha Howell[7], Aaron S. Mansfield[8], Ephraim Parent[9], Manish Thakur[10], Brandon Mancini[11], Jonathan Goldman[12], Jeremie Calais[12], David Bushnell[13], Ye Yuan[14], Joanne Li[14], Nicole Pascale[14], Lucy Gong[14], Denis Ferreira[14], Zhonglin Hao[15]

[1]Huntsman Cancer Center, Salt Lake City, UT; [2]Nebraska Cancer Specialists, Omaha, NE; [3]PanOncology Trials, San Juan, Puerto Rico; [4]Millenium Physicians, Cleveland, TX; [5]Orlando Health Cancer Center, Orlando, FL; [6]University of California, San Francisco, CA; [7]Michigan Health Professionals, Troy, MI; [8]Mayo Clinic, Rochester, MN; [9]Mayo Clinic, Jacksonville, FL; [10]Corewell Health, Grand Rapids, MI; [11]BAMF Health, Grand Rapids, MI; [12]University of California, Los Angeles, CA; [13]University of Iowa, Iowa City, IA; [14]RayzeBio, San Diego, CA; [15]Markey Cancer Center, Lexington, KY

#P1.13A.09

RayzeBio®
A Bristol Myers Squibb Company

BACKGROUND

- Somatostatin receptor type 2 (SSTR2) is expressed in small cell lung cancer (SCLC) in around 48% of samples by IHC[1] and up to 54% of patients with extensive stage disease (ES-SCLC) are positive (>50% lesions positive) on SSTR PET scans.[2]
- RYZ101 (^{225}Ac-DOTATATE) is a first-in-class, highly potent alpha-emitting radiopharmaceutical therapy being developed for SSTR2+ solid tumors (**Figure 1a**):
 - ^{225}Ac has a half life of 9.92 days, and as it decays to stable ^{209}Bi, it generates four short-lived high-energy alpha particles (^{221}Fr, ^{217}At, ^{213}Bi and ^{213}Po) (**Figure 1b**).
 - Alpha-particles have a shorter path length (40–100 µm) and higher linear energy transfer (80–100 keV/µm) than beta-particles (as emitted by ^{177}Lu or ^{90}Y), causing more frequent double-strand DNA breaks and potentially improved therapeutic index (**Figure 1c**).
- RYZ101-101 (NCT05595460) is a single-arm, open-label Phase 1b dose escalation and dose expansion trial of RYZ101 (^{225}Ac-DOTATATE) in combination with 1st line carboplatin + etoposide + atezolizumab in SSTR2+ ES-SCLC patients.[3] Here we present preliminary safety data from Cohort 1.

FIGURE 1. RYZ101 (^{225}Ac-DOTATATE)

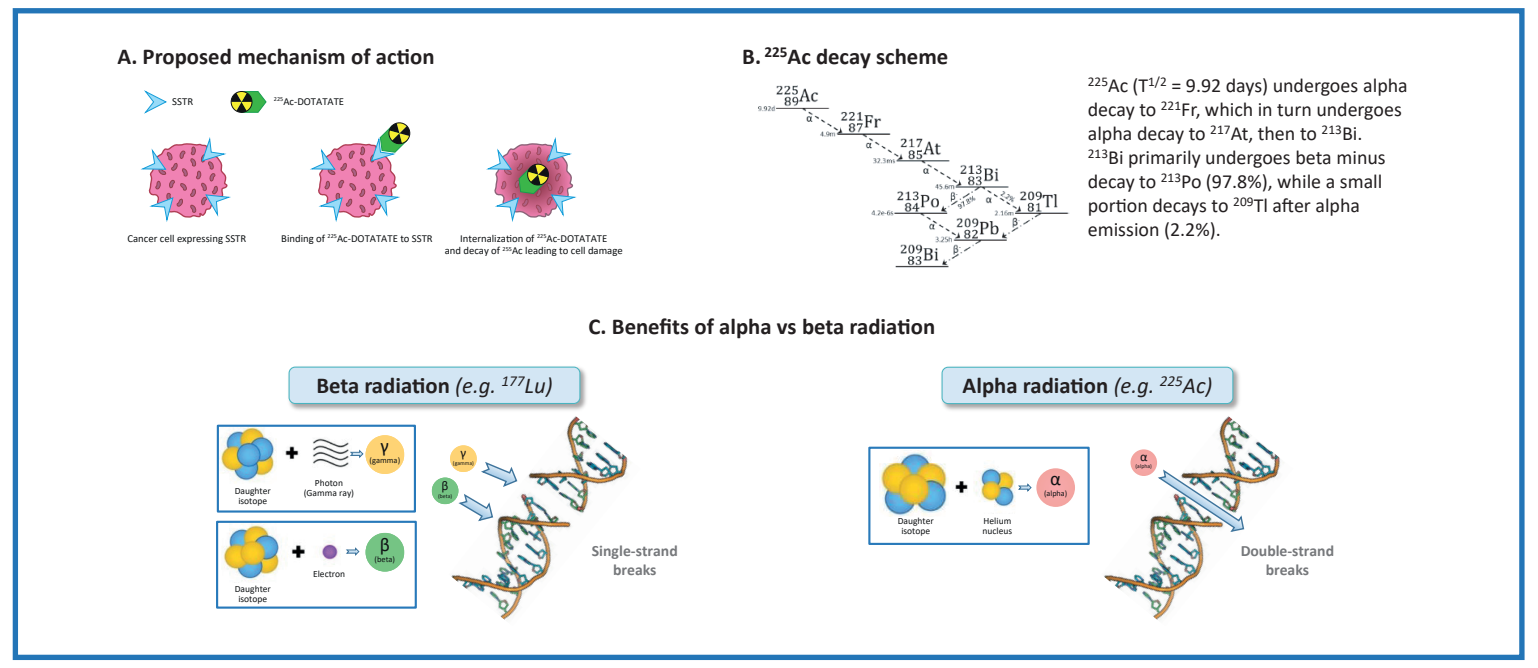
A. Proposed mechanism of action
SSTR
225Ac-DOTATATE
Cancer cell expressing SSTR
Binding of 225Ac-DOTATATE to SSTR
Internalization of 225Ac-DOTATATE and decay of 225Ac leading to cell damage
B. 225Ac decay scheme
225Ac (T1/2 = 9.92 days) undergoes alpha decay to 221Fr, which in turn undergoes alpha decay to 217At, then to 213Bi. 213Bi primarily undergoes beta minus decay to 213Po (97.8%), while a small portion decays to 209Tl after alpha emission (2.2%).
C. Benefits of alpha vs beta radiation
Beta radiation (e.g. 177Lu)
Alpha radiation (e.g. 225Ac)
Daughter isotope
Photon (Gamma ray)
Electron
Helium nucleus
Single-strand breaks
Double-strand breaks

METHODS

- The trial schema, including primary and secondary endpoints, is shown in **Figure 2**.
- Safety analyses:
 - Treatment-emergent adverse events were graded via NCI-CTCAE v5.0.
 - Dose-limiting toxicities (DLTs) were assessed in the first 42 days following the first RYZ101 infusion.
 - Dose escalation decisions and safety data review were overseen by a Data Review Committee (DRC).
 - Tumor assessments via CT/MRI were conducted every 6 weeks following first RYZ101 or SoC infusion with response assessed via RECIST v1.1.
- Pharmacokinetic analysis:
 - Total radioactivity in blood from gamma emissions of ^{225}Ac daughter isotopes were measured during Cycle 1 of RYZ101.
 - Pharmacokinetic parameters were obtained via a noncompartmental analysis of blood radioactive concentrations, decay corrected to sample time.

FIGURE 2. Trial schema

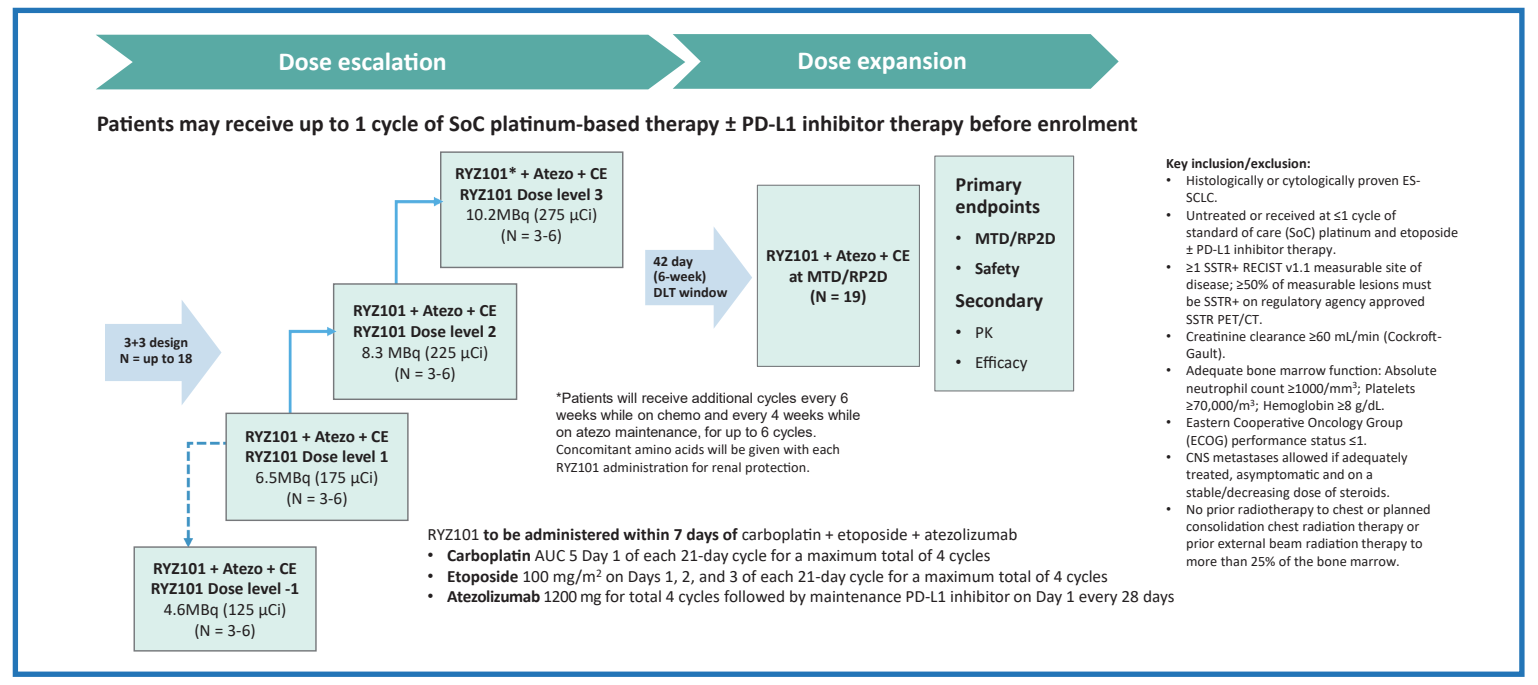
Dose escalation
Dose expansion
Patients may receive up to 1 cycle of SoC platinum-based therapy ± PD-L1 inhibitor therapy before enrolment
3+3 design
N = up to 18
RYZ101 + Atezo + CE
RYZ101 Dose level -1
4.6MBq (125 µCi)
(N = 3-6)
RYZ101 + Atezo + CE
RYZ101 Dose level 1
6.5MBq (175 µCi)
(N = 3-6)
RYZ101 + Atezo + CE
RYZ101 Dose level 2
8.3 MBq (225 µCi)
(N = 3-6)
RYZ101* + Atezo + CE
RYZ101 Dose level 3
10.2MBq (275 µCi)
(N = 3-6)
42 day
(6-week)
DLT window
RYZ101 + Atezo + CE
at MTD/RP2D
(N = 19)
Primary endpoints
MTD/RP2D
Safety
Secondary
PK
Efficacy
*Patients will receive additional cycles every 6 weeks while on chemo and every 4 weeks while on atezo maintenance, for up to 6 cycles. Concomitant amino acids will be given with each RYZ101 administration for renal protection.
RYZ101 to be administered within 7 days of carboplatin + etoposide + atezolizumab
Carboplatin AUC 5 Day 1 of each 21-day cycle for a maximum total of 4 cycles
Etoposide 100 mg/m2 on Days 1, 2, and 3 of each 21-day cycle for a maximum total of 4 cycles
Atezolizumab 1200 mg for total 4 cycles followed by maintenance PD-L1 inhibitor on Day 1 every 28 days
Key inclusion/exclusion:
Histologically or cytologically proven ES-SCLC.
Untreated or received at ≤1 cycle of standard of care (SoC) platinum and etoposide ± PD-L1 inhibitor therapy.
≥1 SSTR+ RECIST v1.1 measurable site of disease; ≥50% of measurable lesions must be SSTR+ on regulatory agency approved SSTR PET/CT.
Creatinine clearance ≥60 mL/min (Cockroft-Gault).
Adequate bone marrow function: Absolute neutrophil count ≥1000/mm3; Platelets ≥70,000/m3; Hemoglobin ≥8 g/dL.
Eastern Cooperative Oncology Group (ECOG) performance status ≤1.
CNS metastases allowed if adequately treated, asymptomatic and on a stable/decreasing dose of steroids.
No prior radiotherapy to chest or planned consolidation chest radiation therapy or prior external beam radiation therapy to more than 25% of the bone marrow.

Atezo, atezolizumab; BM, bone marrow; CE, carboplatin + etoposide; ES-SCLC, extensive stage small cell lung cancer (Stage IV (T any, N any, M 1a/b/c) or T 3-4 due to multiple lung nodules or tumor/nodal volume that is not amenable to a tolerable radiation plan); MTD, maximum tolerated dose; PK, pharmacokinetics; RP2D, recommended Phase 2 dose; SSTR, somatostatin receptor

RESULTS

Status and baseline demographics

- As of the data cutoff of 13-May-2024:
 - Four patients have been enrolled and received at least one dose of RYZ101:
 - Two patients completed all 4 cycles of SoC.
 - One patient completed all 6 cycles of RYZ101 and continues on atezolizumab maintenance therapy and is in long-term follow up.
 - Enrollment into Cohort 1 dose level 1 (6.5 MBq) has been completed.
 - Baseline demographics are shown in **Table 1**.

Safety

- A summary of adverse events is listed in **Table 2**:
 - All patients in Cohort 1 experienced a treatment-emergent adverse event (TEAE).
 - No dose-limiting toxicities (DLTs) were reported in the three DLT-evaluable patients. The fourth patient was not evaluable for DLTs as they stopped treatment early due to progressive disease.
 - The majority of RYZ101 treatment-related adverse events (AEs) were low grade with one Grade 4 AE (lymphocyte count decreased) This event was not considered to be a DLT (per protocol definition).
 - One patient required dose reduction of RYZ101 for unrelated Grade 4 neutropenia and one subject had an interruption of RYZ101 for unrelated Grade 3 syncope.
 - No serious AEs (SAEs) related to RYZ101 were reported.
 - Three patients were hospitalized due to TEAEs that were considered related to the underlying disease and not related to either SoC or RYZ101.
- All TEAEs experienced by patients in Cohort 1 are listed in **Table 3**.
- Grade 3 or higher TEAEs are listed in **Table 4**:
 - All were Grade 3 except for decreased neutrophil count, neutropenia, lymphocyte count decreased and spinal cord compression.
 - All Grade 3 or higher TEAEs were unrelated to RYZ101 except for Grade 4 Lymphocyte count decreased.
- SAEs are listed in **Table 5**:
 - Three patients in Cohort 1 experienced an SAE.
 - All SAEs were considered unrelated to RYZ101.
- Adverse events of special interest (AESIs) are listed in **Table 6**:
 - AESIs are defined as Grade 3 or higher thrombocytopenia, Grade 3/4 any other hematotoxicity (except lymphocytopenia), secondary hematologic malignancies and nephrotoxicity.
 - Hematotoxicity was the only AESI noted in Cohort 1 with highest grade being Grade 4 neutropenia and decreased neutrophils.
- Laboratory value changes from baseline did not worsen to Grade 3 or higher for hemoglobin, platelets or creatinine clearance. One patient had a worsening of neutrophil count from baseline to Grade 4.
- One patient in Cohort 1 remains on atezolizumab maintenance therapy without significant toxicities.

Pharmacokinetics

- Mean volume of distribution was 1.69 L/kg (CV 90.6%), mean clearance was 1.72 mL/min/kg (coefficient of variation 88.2%) and terminal half-life was 62.7 hours.

TABLE 1. Patient demographics

Category	Cohort 1: 6.5 MBq (N=4)
Age, years	
Median (range)	63.3 (49, 79)
Age group, n (%)	
<65 years	2 (50)
≥65 years	2 (50)
≥75 years	1 (25)
Sex, n (%)	
Male	2 (50)
Female	2 (50)
Race, n (%)	
White	4 (100)
ECOG Performance Status at baseline, n (%)	
0	2 (50.0)
1	2 (50.0)
Ever smoked/used tobacco	
Yes	4 (100)
Cycle 1 SoC during screening	4 (100)

Data cutoff 13 May, 2024 ECOG, Eastern Cooperative Oncology Group; SoC, standard of care

TABLE 2. Summary of adverse events

Variable/category	Cohort 1: 6.5 MBq (N=4)
Any TEAE, n (%)	4 (100)
Any serious AE, n (%)	3 (75)
Any treatment-related TEAE, n (%)	3 (75)
RYZ101	3 (75)
SoC	3 (75)
Any treatment-related SAE, n (%)	0
Any Grade 3 or 4 TEAE, n (%)	4 (100)
Any fatal TEAE (Grade 5), n (%)	0
Any treatment-related Grade 3 or 4 TEAE, n (%)	3 (75)
RYZ101	1 (25)
SoC	2 (50)
Any treatment-related Grade 3 or 4 SAE, n (%)	0
Dose-limiting toxicities,[a] n (%)	0
TEAEs leading to hospitalization, n (%)	3 (75)
TEAEs leading to discontinuation, n (%)	0
TEAEs leading to dose holds/modifications/delays, n (%)	2 (50)

[a] The DLT-evaluable population includes any patient who received at least 1 dose of RYZ101 and completed the 42-day DLT review period or experienced a DLT. Three patients were DLT-evaluable and one patient was not considered DLT-evaluable as they stopped treatment early due to progressive disease during the DLT period and went into hospice care
AE, adverse event; SAE, serious adverse event; SoC, standard of care; TEAE, treatment-emergent adverse event

TABLE 3. Treatment-emergent adverse events

Preferred AE term	Cohort 1: 6.5 MBq (N=4) n (%)	AE term	Cohort 1: 6.5 MBq (N=4) n (%)
Decreased neutrophils	2 (50.0)	Hypokalemia	1 (25.0)
Hyponatremia	2 (50.0)	Hypothyroidism	1 (25.0)
Urinary tract infection	2 (50.0)	Influenze-like illness	1 (25.0)
Constipation	2 (50.0)	Leukopenia	1 (25.0)
Spinal fracture	2 (50.0)	Lymph count decreased	1 (25.0)
Anemia	1 (25.0)	Nausea	1 (25.0)
Atrial fibrillation	1 (25.0)	Neutropenia	1 (25.0)
Back pain	1 (25.0)	Pain	1 (25.0)
TSH increased	1 (25.0)	Platelet count decreased	1 (25.0)
Chills	1 (25.0)	Pleural effusion	1 (25.0)
Decreased appetite	1 (25.0)	Pyrexia	1 (25.0)
Dehydration	1 (25.0)	Spinal cord compression	1 (25.0)
Diarrhea	1 (25.0)	Syncope	1 (25.0)
Dizziness	1 (25.0)	Thrombocytopenia	1 (25.0)
Eosinophilia	1 (25.0)	Tremor	1 (25.0)
Fatigue	1 (25.0)	Vomiting	1 (25.0)
Hot flush	1 (25.0)	Weight decreased	1 (25.0)
Hypercalcemia	1 (25.0)	WBC count decreased	1 (25.0)
Hypertension	1 (25.0)		

AE, adverse event; WBC, white blood cell

REFERENCES

1. Lehman JM, et al. Int J Cancer 2019;144:1104–14.
2. Sen F, et al. Cancers (Basel) 2023;15:3595.
3. ClinicalTrials.gov. Available at: https://clinicaltrials.gov/study/NCT05595460

TABLE 4. Grade 3 or higher treatment-emergent adverse events

Variable/category	Cohort 1: 6.5 MBq (N=4)
Any Grade 3 or higher TEAE, n (%)	4 (100)
Nausea	1 (25)
Pain	1 (25)
Back pain	1 (25)
Decreased neutrophils	1 (25)
Neutropenia	1 (25)
Decreased white blood cell count	1 (25)
Leukopenia	1 (25)
Lymphocyte count decreased	1 (25)
Hyponatremia	2 (50)
Syncope	1 (25)
Spinal cord compression	1 (25)
Hypertension	1 (25)

TEAE, treatment-emergent adverse event

TABLE 5. Serious adverse events

Category	Cohort 1: 6.5 MBq (N=4)
Any SAE, n (%)	3 (75)
Pain	1 (25)
Hyponatremia	1 (25)
Back pain	1 (25)
Syncope	1 (25)
Spinal cord compression	1 (25)

SAE, serious adverse event

TABLE 6. Adverse events of special interest

Category	Cohort 1: 6.5 MBq (N=4)
Any AESI, n (%)	1 (25)
Hematotoxicity	1 (25)
Max Grade = 4	
Decreased neutrophils	1 (25)
White blood cell count decrease	1 (25)
Leukopenia	1 (25)
Neutropenia	1 (25)

AESI, adverse event of special interest

CONCLUSIONS

- Preliminary data show that the safety profile of RYZ101 in combination with chemoimmunotherapy is acceptable and manageable. There were no DLTs reported, and no SAEs were deemed related to RYZ101.
- Preliminary pharmacokinetic findings indicate clearance and half-life comparable to previous studies of RYZ101 and ^{177}Lu-DOTATATE.
- Data from Cohort 1 Dose Level 1 (6.5 MBq) was reviewed by the DRC with recommendation to continue escalation to higher dose levels.

ACKNOWLEDGMENTS

- The authors would like to thank all patients and their caregivers, and all site investigators and study staff for participating in this study.
- The ^{225}Ac used in this research was supplied by multiple sources, including the U.S. Department of Energy Isotope Program managed by the Office of Isotope R&D and Production.
- The study is sponsored by RayzeBio Inc., San Diego, CA, USA. The study sponsor also funded medical editing/writing support for this poster, which was provided by Miller Medical Communications Ltd.

Presented at the IASLC World Conference on Lung Cancer, San Diego, CA, USA, September 7–10, 2024